A Novel Product Candidate (CPTX2309) for In Vivo mRNA Engineering of Anti-CD19 CAR T Cells Utilizing Novel CD8-Targeted Lipid Nanoparticles

Theresa L. Hunter[1], Ferran Soldevila[1], Yan Zhang[1], Brittany Ross[1], Daiki Matsuda[1], Yanjie (Belle) Bao[1], John J. Li[1], Michelle Nguyen[1], Matthew Butcher[1], Michael Pica[1], Claudia Fernandez[1], James Vestal[1], Goutam Mondal[1], Yi Kuo[1], Jeffrey Chen[1], Josephine Nguyen[1], Young Yoon Choi[1], Diana Galvan[1], Duy P. Nguyen[1], Donald Jhung[1], Stuart A. Sievers[1], Steven P. Tanis[1], Cory Bentley[1], Michael Rosenzweig[1], Priya Karmali[1], Adrian I. Bot[1], Haig Aghajanian[1], Gregor B. Adams[1]

[1]Capstan Therapeutics, Inc.; San Diego, California, USA.

BACKGROUND

Autologous chimeric antigen receptor (CAR) T cell therapies have revolutionized cancer treatment and are now demonstrating promising activity in a number of autoimmune diseases (AID). Challenges in cell manufacturing, scaling, and the need for inpatient treatment highlight the necessity for an off-the-shelf solution for AID, devoid of lymphodepletion conditioning and viral vectors. Inspired by the success of CAR products and of mRNA lipid nanoparticles (LNPs) in COVID vaccines, Capstan is developing CPTX2309, a product of our CellSeeker™ tLNP platform, to deliver a sequence-enhanced mRNA payload encoding an anti-CD19 CAR to preferentially reprogram CD8-expressing T cells *in vivo* for treating AID.

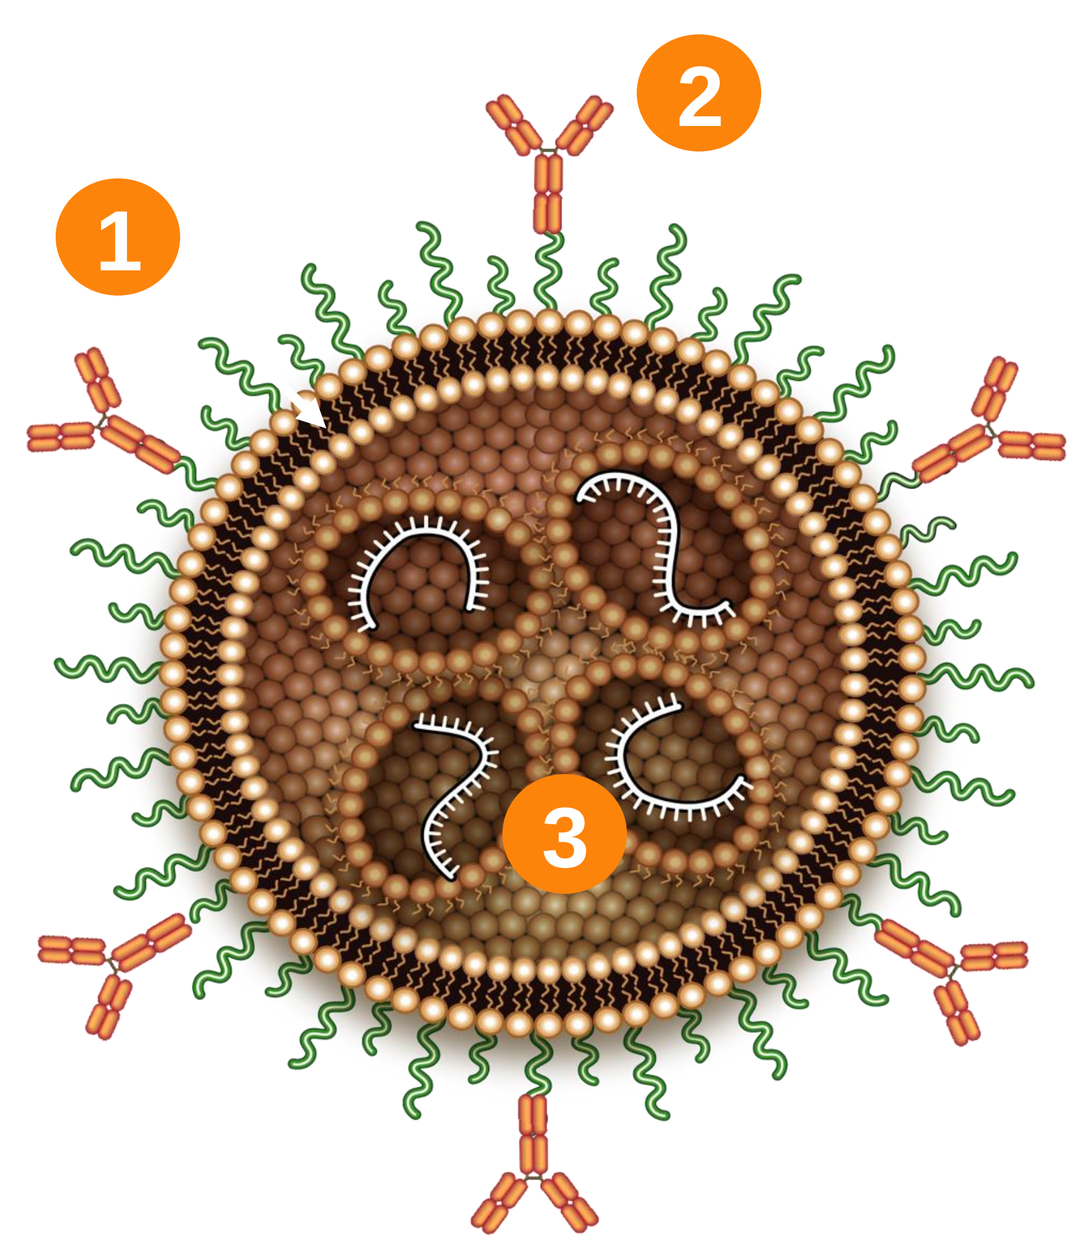
1
2
3

CPTX2309 IS MADE UP OF THREE COMPONENTS:

1. LNP DELIVERY VEHICLE

Capstan's proprietary lipid nanoparticle is a non-viral dose-tunable system designed for increased tolerability and biodegradability to allow for repeat *in vivo* dosing.

2. CD8 TARGETING BINDERS

Humanized CD8 antibody conjugated onto the nanoparticle surface, creating targeted lipid nanoparticles (tLNPs) to enable engineering of CD8 T cells and for preferential delivery to specific cells of disease-specific payloads.

3. ENHANCED ANTI-CD19 CAR mRNA

mRNA with improved performance encoding a fully human CAR recognizing CD19.

FIGURE 1 | DOSE DEPENDENT ENGINEERING OF NON-ACTIVATED HUMAN CD8 T CELLS USING CPTX2309

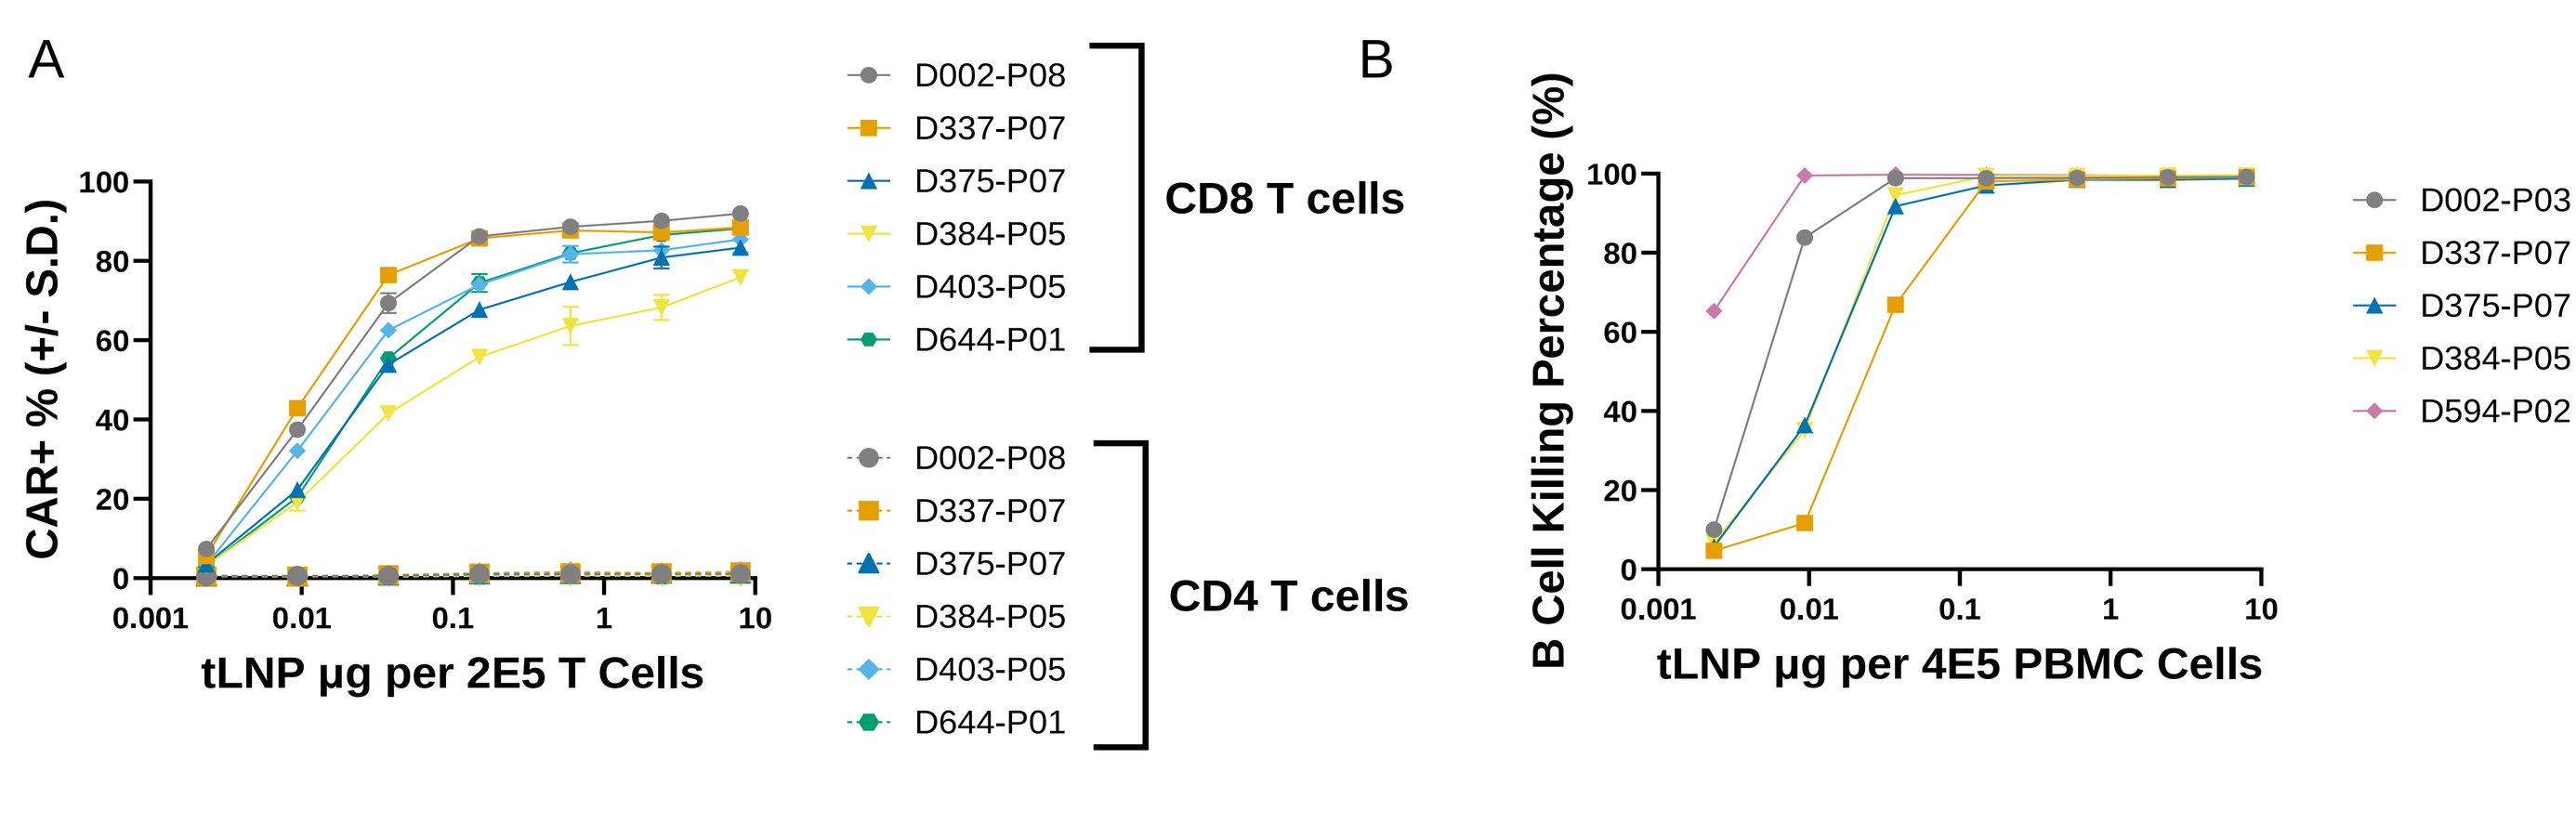
A
CAR+ % (+/- S.D.)
tLNP µg per 2E5 T Cells
D002-P08
D337-P07
D375-P07
D384-P05
D403-P05
D644-P01
CD8 T cells
CD4 T cells
B
B Cell Killing Percentage (%)
tLNP µg per 4E5 PBMC Cells
D002-P03
D337-P07
D375-P07
D384-P05
D594-P02

A) Percent CAR expression 24 hours after *in vitro* transfection of non-activated human T cells with CPTX2309. B) Killing of autologous B cells 72 hours after *in vitro* transfection of non-activated human T cells with CPTX2309. Killing measured by flow cytometry and normalized to non-transfected control. Each color represents a separate human donor.

FIGURE 2 | CPTX2309 ENGINEERED NON-ACTIVATED CD8 T CELLS DEMONSTRATE ANTIGEN-SPECIFIC FUNCTION

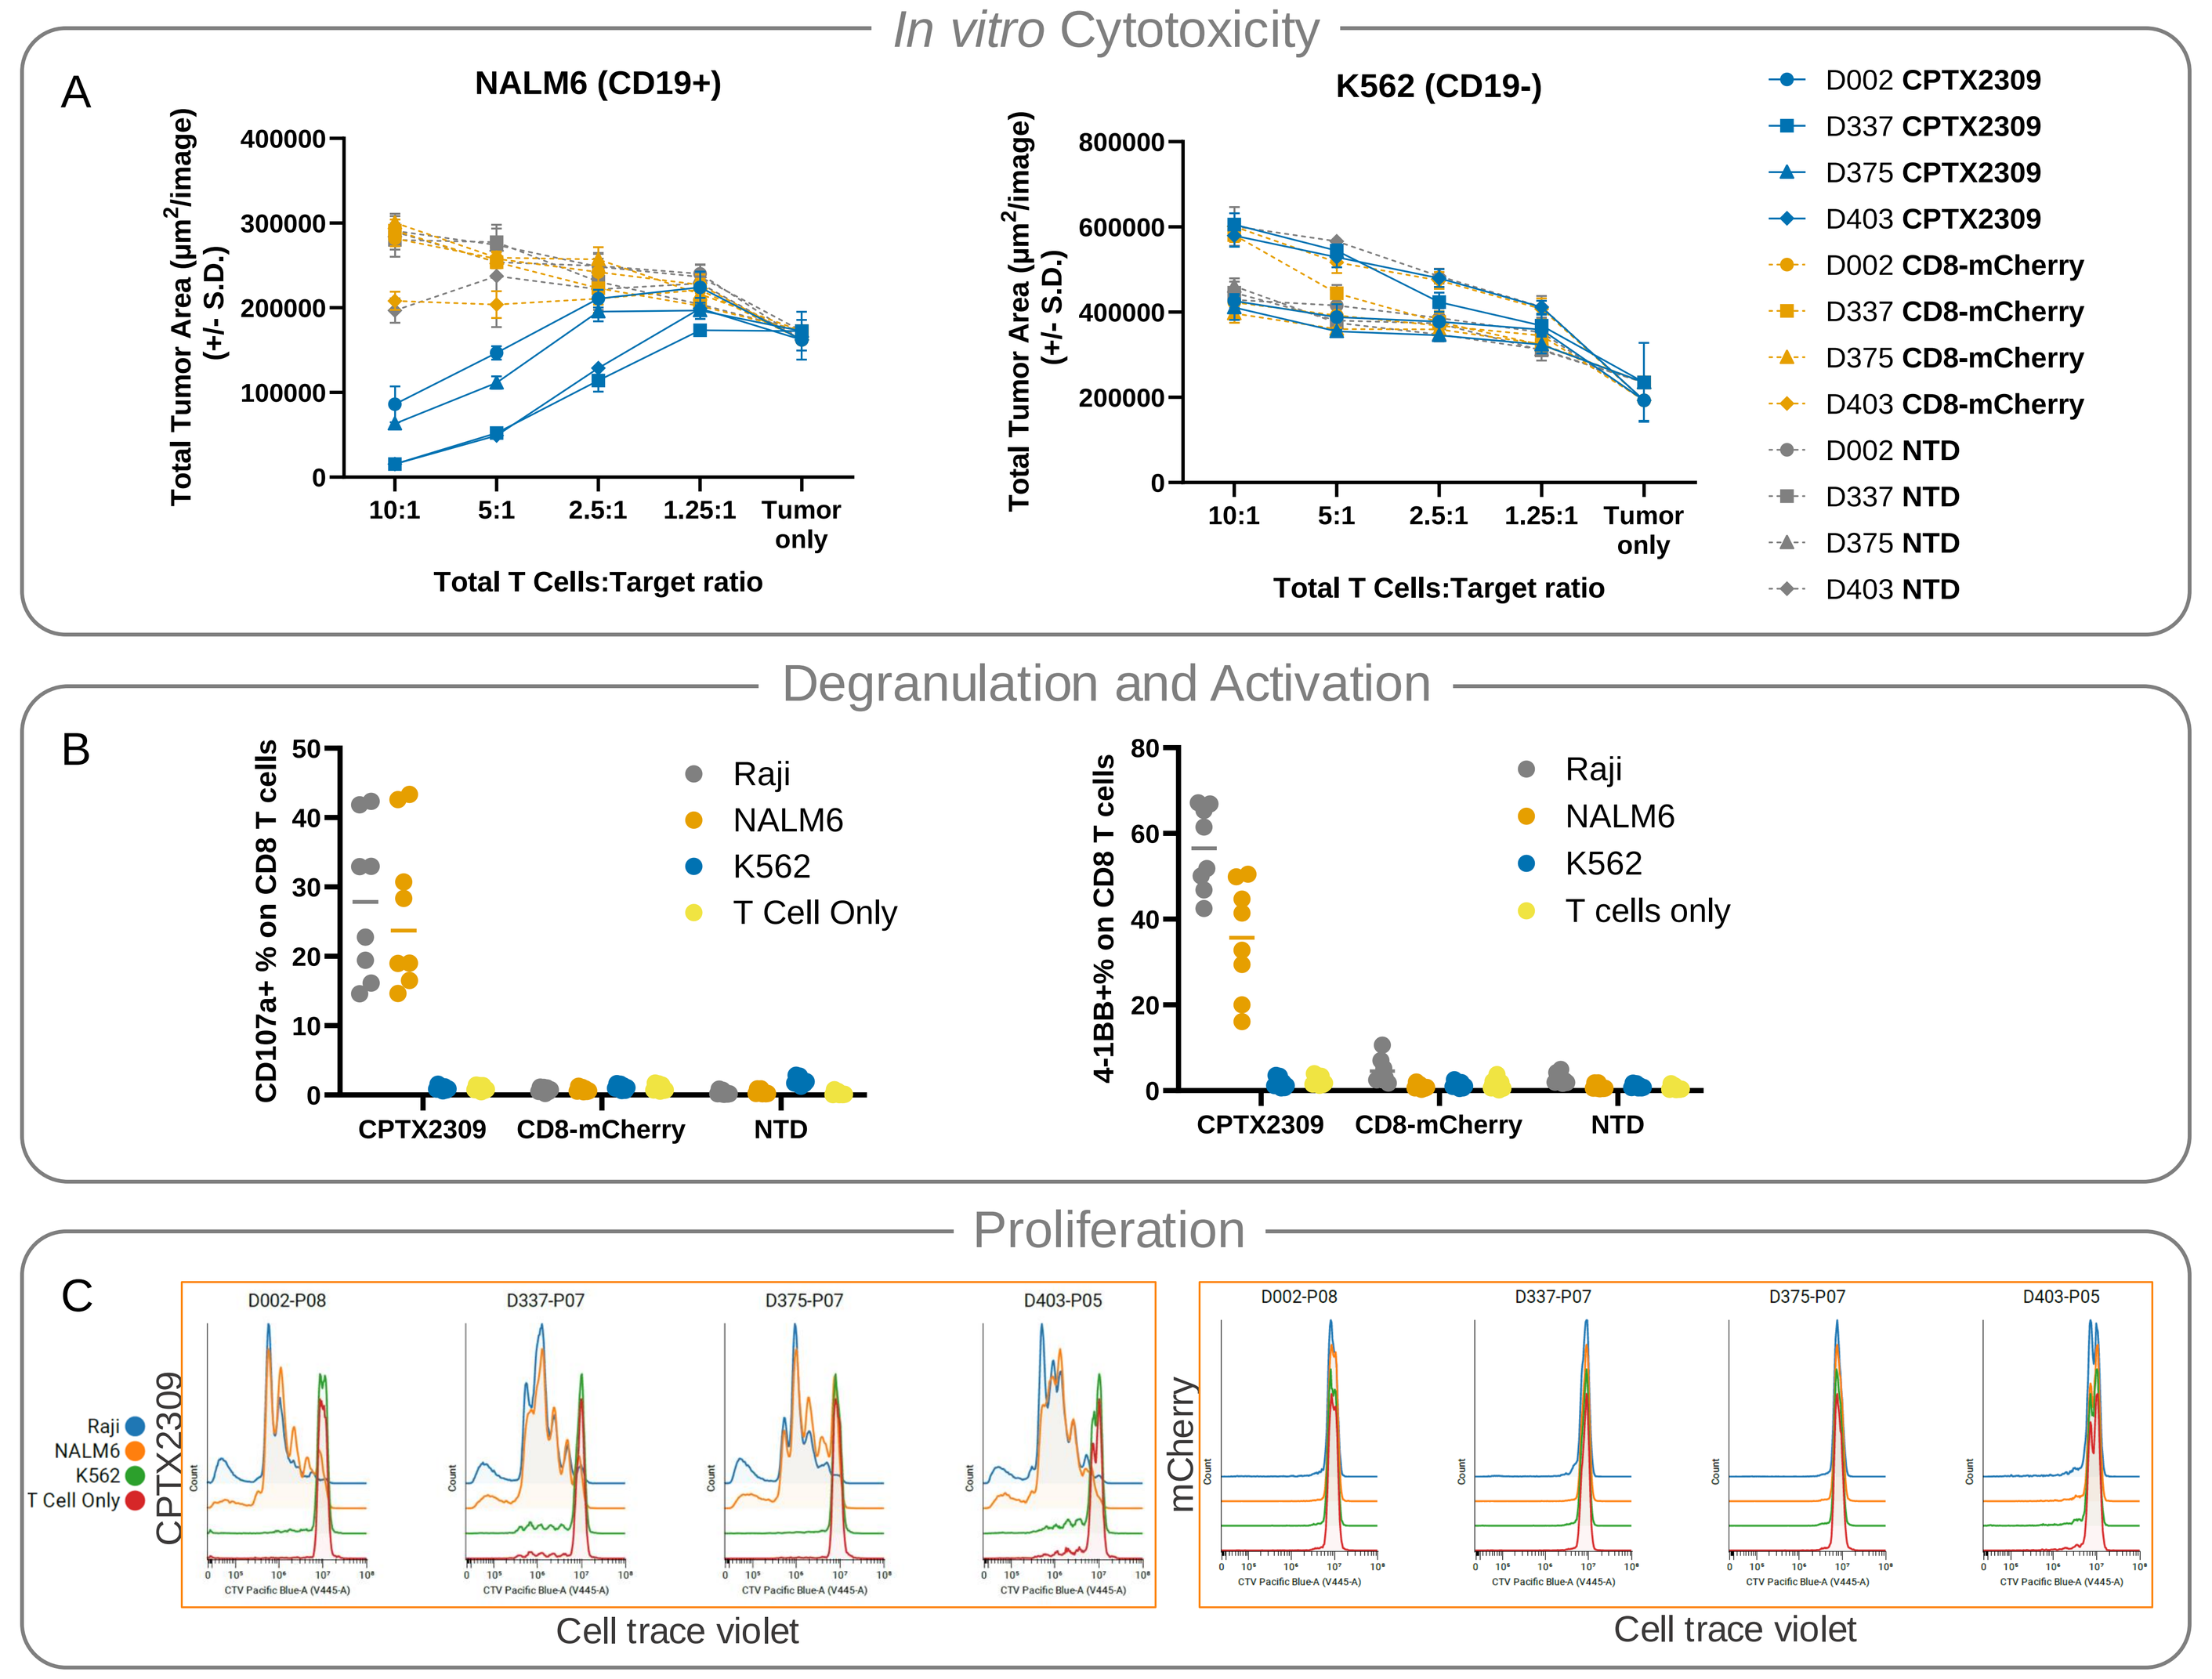
In vitro Cytotoxicity
A
NALM6 (CD19+)
K562 (CD19-)
Total Tumor Area (µm²/image) (+/- S.D.)
Total T Cells:Target ratio
10:1
5:1
2.5:1
1.25:1
Tumor only
D002 CPTX2309
D337 CPTX2309
D375 CPTX2309
D403 CPTX2309
D002 CD8-mCherry
D337 CD8-mCherry
D375 CD8-mCherry
D403 CD8-mCherry
D002 NTD
D337 NTD
D375 NTD
D403 NTD
Degranulation and Activation
B
CD107a+ % on CD8 T cells
4-1BB+% on CD8 T cells
Raji
NALM6
K562
T Cell Only
T cells only
CPTX2309
CD8-mCherry
NTD
Proliferation
C
CPTX2309
mCherry
D002-P08
D337-P07
D375-P07
D403-P05
Cell trace violet

Non-activated T cells were transfected with CPTX2309 or mCherry control. A) Cytotoxicity against CD19+ (Nalm6, left) and CD19- (K562, right) target cells. NTD = non-transfected control.
B) Degranulation measured by surface CD107a (left) and 4-1BB expression (right) after co-culture with CD19+ (Raji, Nalm6) or CD19- (K562) tumor cells. C) Proliferation of CPTX2309 (left) or mCherry (right) transfected T cells after co-culture with indicated tumor cells. Data is showing CD8 T cell proliferation with no proliferation observed in CD4 T cells (data not shown).

FIGURE 3 | CPTX2309 ENGINEERED CAR T CELLS ARE POLYFUNCTIONAL

Cytokine Production in CPTX2309-Transfected T cells upon Co-Culture with NALM6 (pg/mL)

Payload	E:T	IFN-gamma	TNF-alpha	GM-CSF	IL-6	IL-8	IP-10	MCP-1	Rantes
CPTX2309	9:1	2,057	41	2,135	44	346	25	16	686
	3:1	1,576	34	2,025	19	245	16	21	856
	1:1	713	22	868	11	110	8	15	458
	0.3:1	197	11	196	0	36	4	19	181
CD8-mCherry	9:1	520	38	695	44	365	21	6	484
	3:1	196	13	240	13	65	9	9	188
	1:1	58	5	42	0	23	2	10	68
	0.3:1	11	0	7	0	7	2	15	46
NTD	9:1	493	40	956	36	422	24	23	234
	3:1	121	11	140	11	39	5	11	78
	1:1	62	0	93	6	29	4	16	73
	0.3:1	9	0	16	0	4	4	2	37

Cytokine Production in CPTX2309-Transfected PBMC (pg/mL)

Ctn Id	TLNP dose	IFN-gamma	TNF-alpha	GM-CSF	IL-6	IL-8	IP-10	MCP-1	RANTES	IFN-B
CPTX2309	0.6ug	19,398	1,131	1,507	10,534	53,971	43,240	44,783	727	469
	0.0375..	6,513	621	1,567	2,878	47,756	56,196	44,943	513	483
	0.01ug	3,389	364	1,654	1,411	66,983	27,127	43,808	341	403
CD8-Luciferase	0.6ug	204	248	444	9,345	24,626	9,385	24,806	134	185
	0.0375..	136	35	456	194	3,508	11,728	4,522	110	65
	0.01ug	66	65	345	727	12,444	8,175	9,988	107	83
NTD	0.6ug	31	12	149	88	2,605	3,432	642	80	54
	0.0375..	34	11	211	62	1,513	4,156	290	86	96
	0.01ug	27	22	195	70	1,725	4,208	327	79	49

Secreted cytokines were measured after co-culture of human T cells transfected with CPTX2309 or control with CD19 expressing NALM6 tumor cells at four total T cells : target (E:T) ratios (top) or cytokine production in PBMC upon transfection with CPTX2309 (which transfects CD8 T cells to mediate B cell killing) or control at 3 tLNP doses (bottom). NTD: Non-transfected control.

FIGURE 4 | RAPID TARGET CELL CLEARANCE USING tLNP mRNA CAR-ENGINEERED CELLS *IN VITRO*

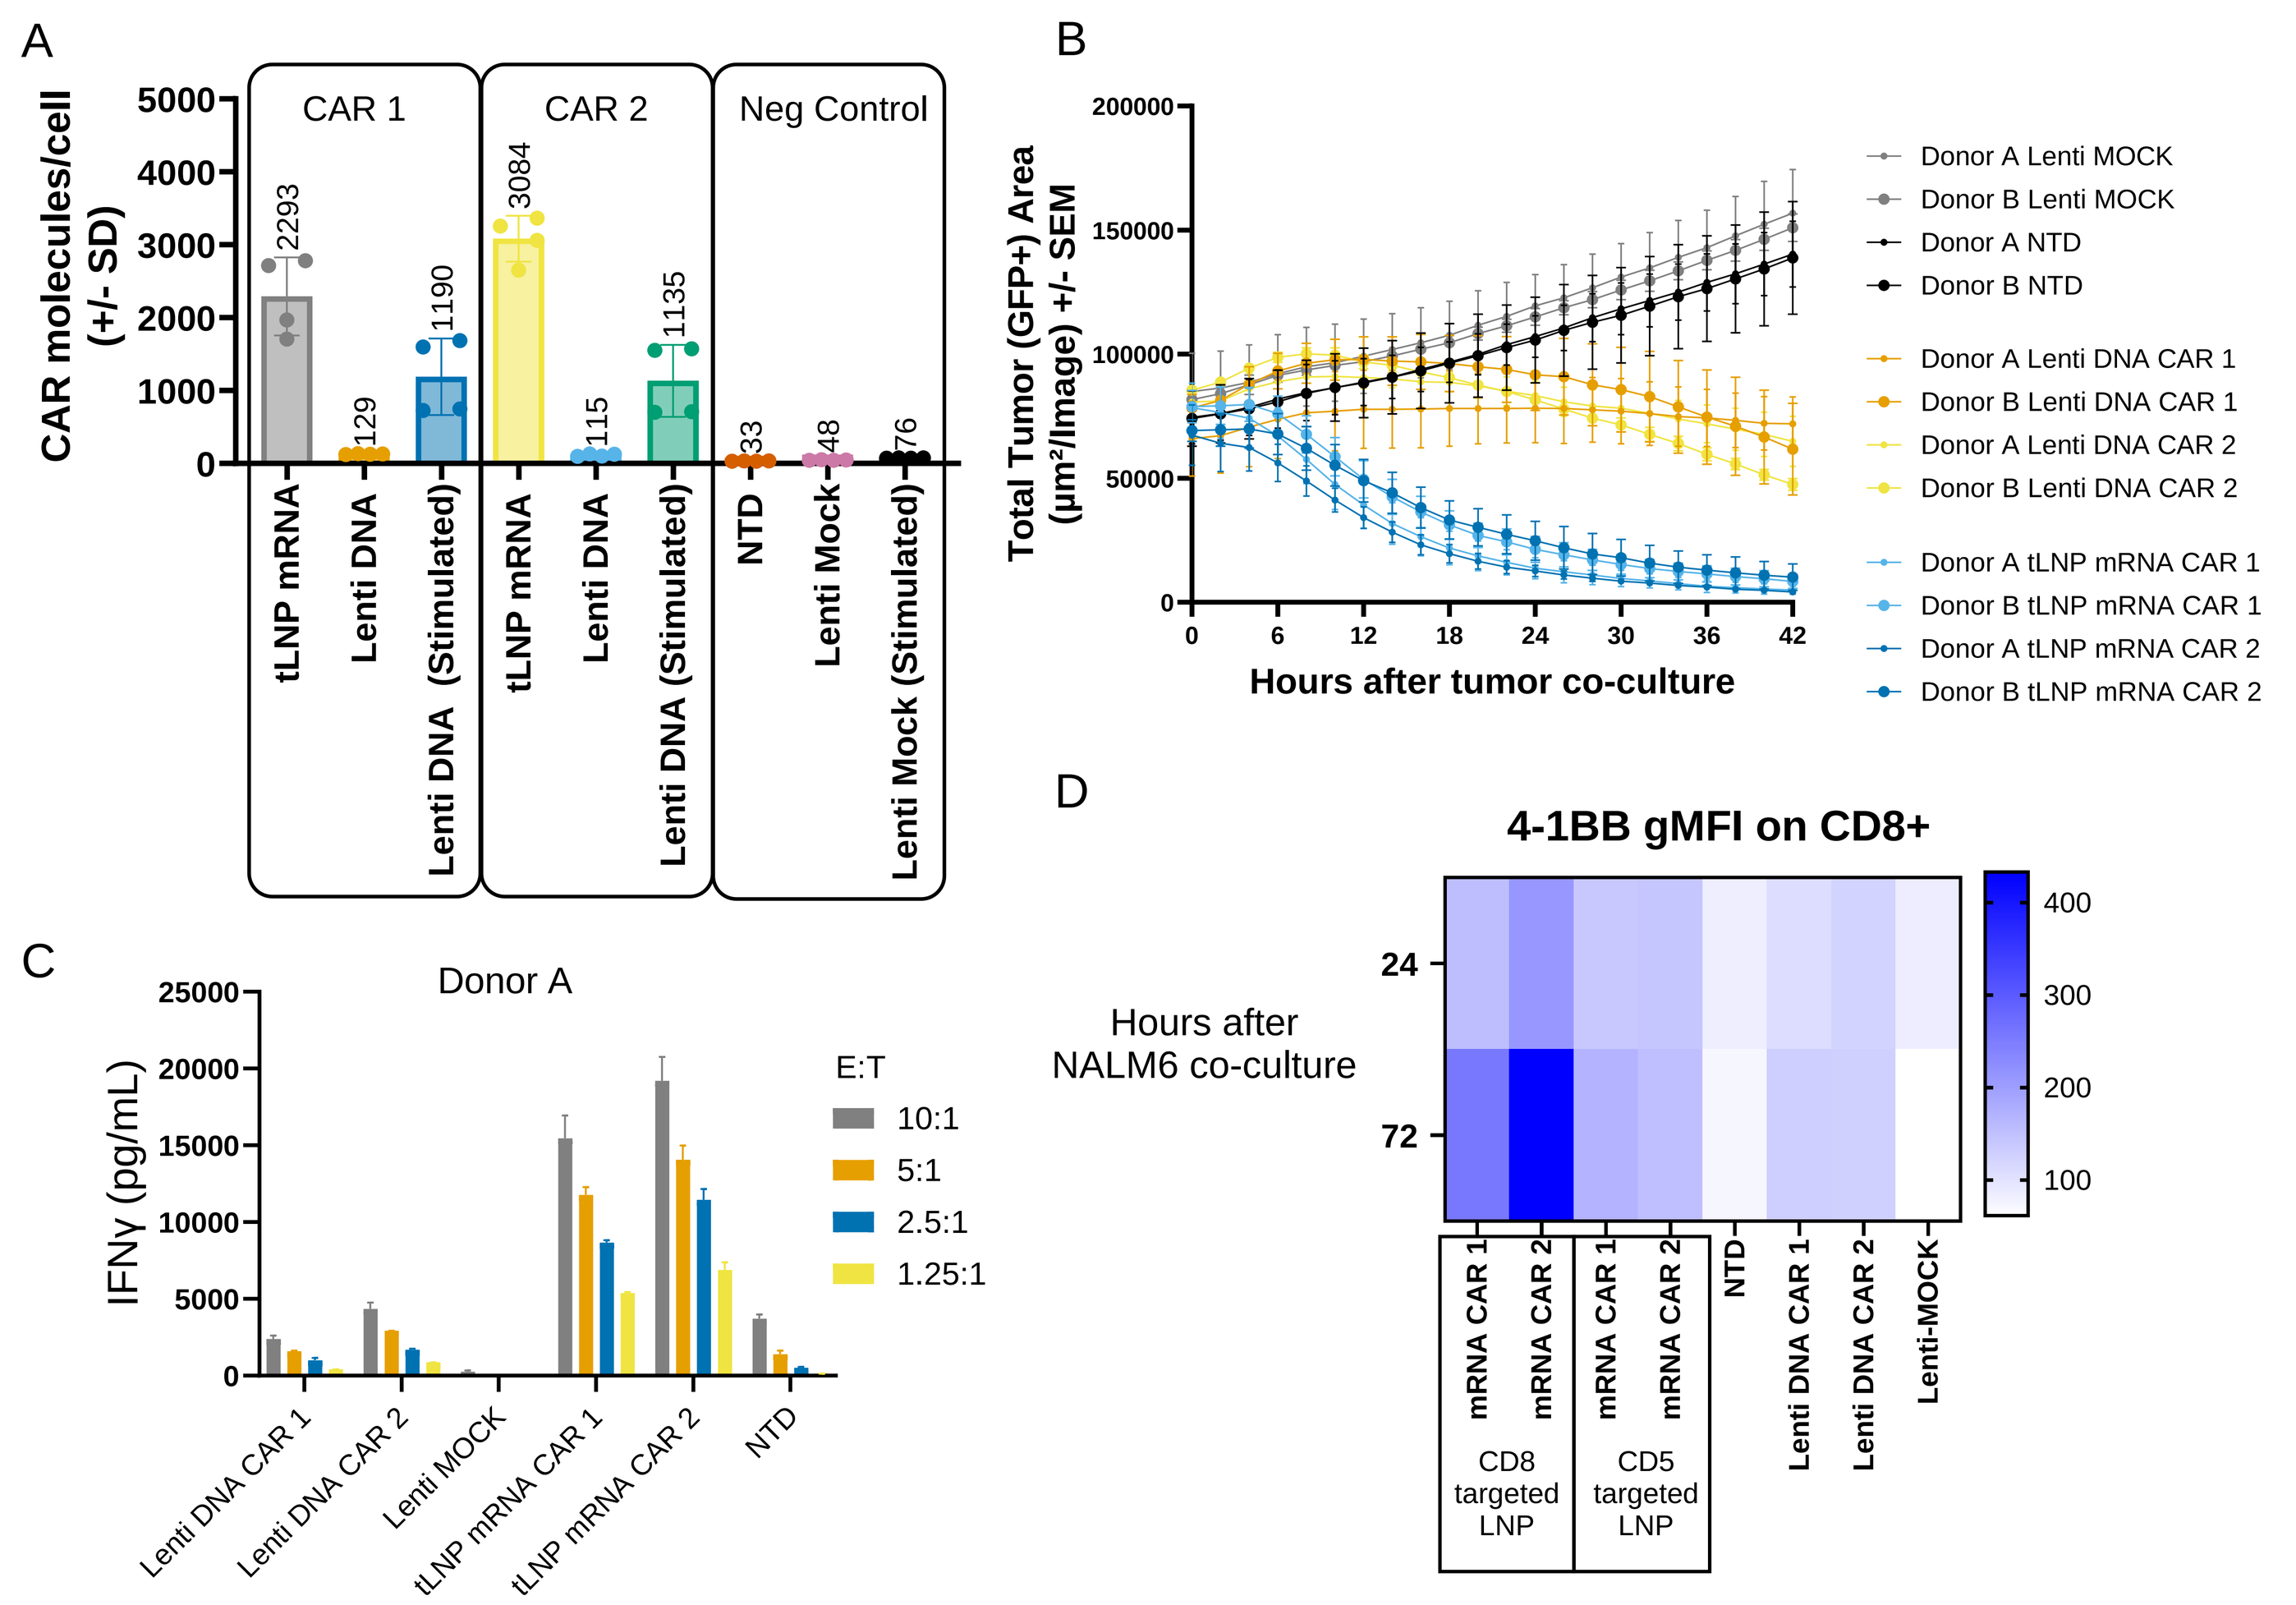
A
CAR molecules/cell (+/- SD)
CAR 1
CAR 2
Neg Control
2293
129
1190
3084
115
1135
33
48
76
tLNP mRNA
Lenti DNA
Lenti DNA (Stimulated)
NTD
Lenti Mock
Lenti Mock (Stimulated)
B
Total Tumor (GFP+) Area (µm²/image) +/- SEM
Hours after tumor co-culture
Donor A Lenti MOCK
Donor B Lenti MOCK
Donor A NTD
Donor B NTD
Donor A Lenti DNA CAR 1
Donor B Lenti DNA CAR 1
Donor A Lenti DNA CAR 2
Donor B Lenti DNA CAR 2
Donor A tLNP mRNA CAR 1
Donor B tLNP mRNA CAR 1
Donor A tLNP mRNA CAR 2
Donor B tLNP mRNA CAR 2
C
Donor A
IFNγ (pg/mL)
E:T
10:1
5:1
2.5:1
1.25:1
Lenti DNA CAR 1
Lenti DNA CAR 2
Lenti MOCK
tLNP mRNA CAR 1
tLNP mRNA CAR 2
NTD
D
4-1BB gMFI on CD8+
Hours after NALM6 co-culture
mRNA CAR 1
mRNA CAR 2
CD8 targeted LNP
CD5 targeted LNP
Lenti DNA CAR 1
Lenti DNA CAR 2
Lenti-MOCK

A) CAR molecules per cell quantified using PE Quantibrite beads (BD) on T cells transfected with tLNP containing mRNA CAR payload, T cells transduced with DNA lentiCAR, or negative controls. mRNA CAR expression was detected at 24 hours after transfection and Lenti DNA CAR expression was measured post cell thaw with (stimulated) or without dynabeads stimulation for 3 days. B) GFP-expressing tumor (target) cell killing by tLNP-transfected or lentiCAR-transduced T cells as measured by Incucyte imaging of GFP area. C) IFNγ produced by tLNP-transfected or lentiCAR-transduced T cells after co-culture with CD19+ tumor cells (representative data from one donor). A-B-C: CD5 targeted LNP was used to deliver mRNA CAR payload into both CD4 and CD8 T cells as lenti DNA CAR are CAR+ in both CD4 and CD8 cells. D) 4-1BB expression measured on tLNP-transfected or lentiCAR-transduced T cells at 24 and 72 hours after co-culture with CD19+ NALM6 cells. mRNA CAR and lenti DNA CAR have the same CAR amino acid sequence. CAR2 is the same CAR sequence in CPTX2309. NTD: non-transfected control.

FIGURE 5 | RAPID B-CELL TARGET CLEARANCE USING CPTX2309 *IN VIVO* AFTER A SINGLE DOSE

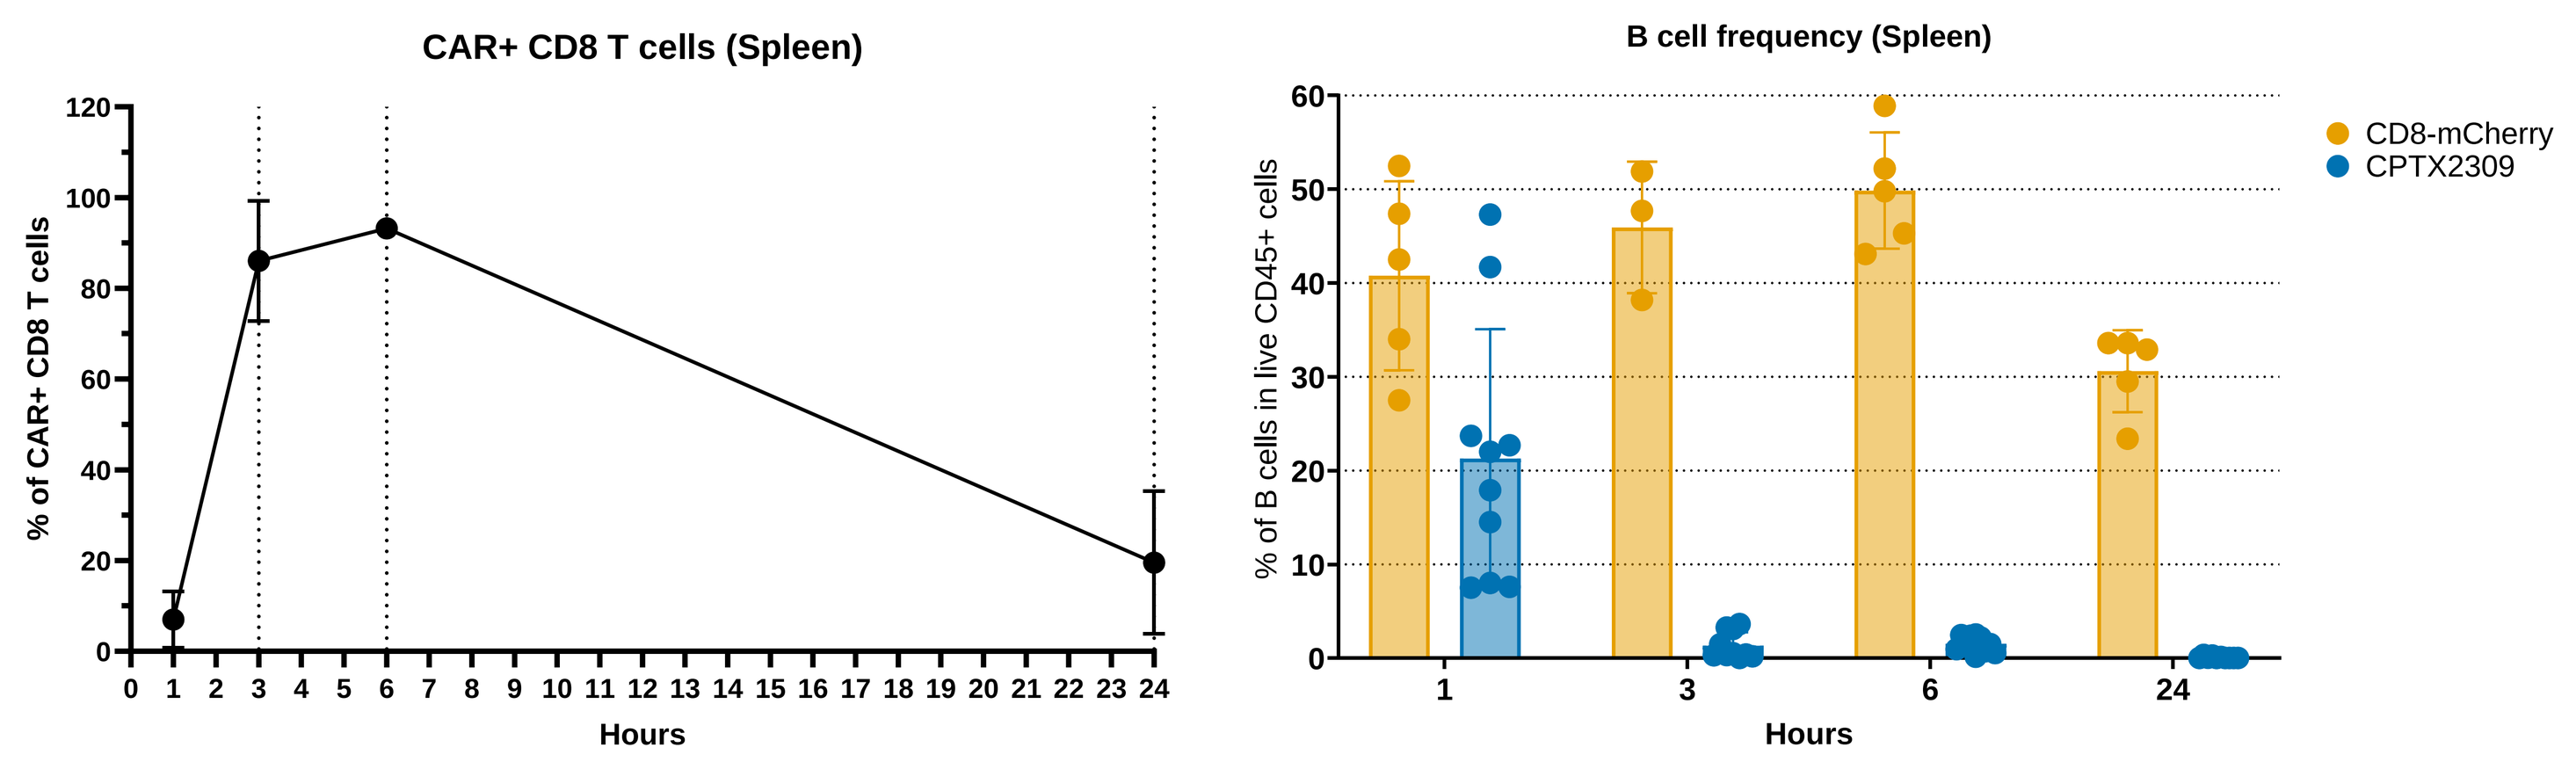
CAR+ CD8 T cells (Spleen)
% of CAR+ CD8 T cells
Hours
B cell frequency (Spleen)
% of B cells in live CD45+ cells
CD8-mCherry
CPTX2309

Frequencies of CAR expression (left) and B cells (right) in NSG mice engrafted with human PBMCs and treated with 30 µg of CPTX2309 or CD8-mCherry control.

FIGURE 6 | MINIMAL tLNP-CAR DOSE NEEDED TO ELIMINATE HUMAN PRIMARY B CELLS *IN VIVO*

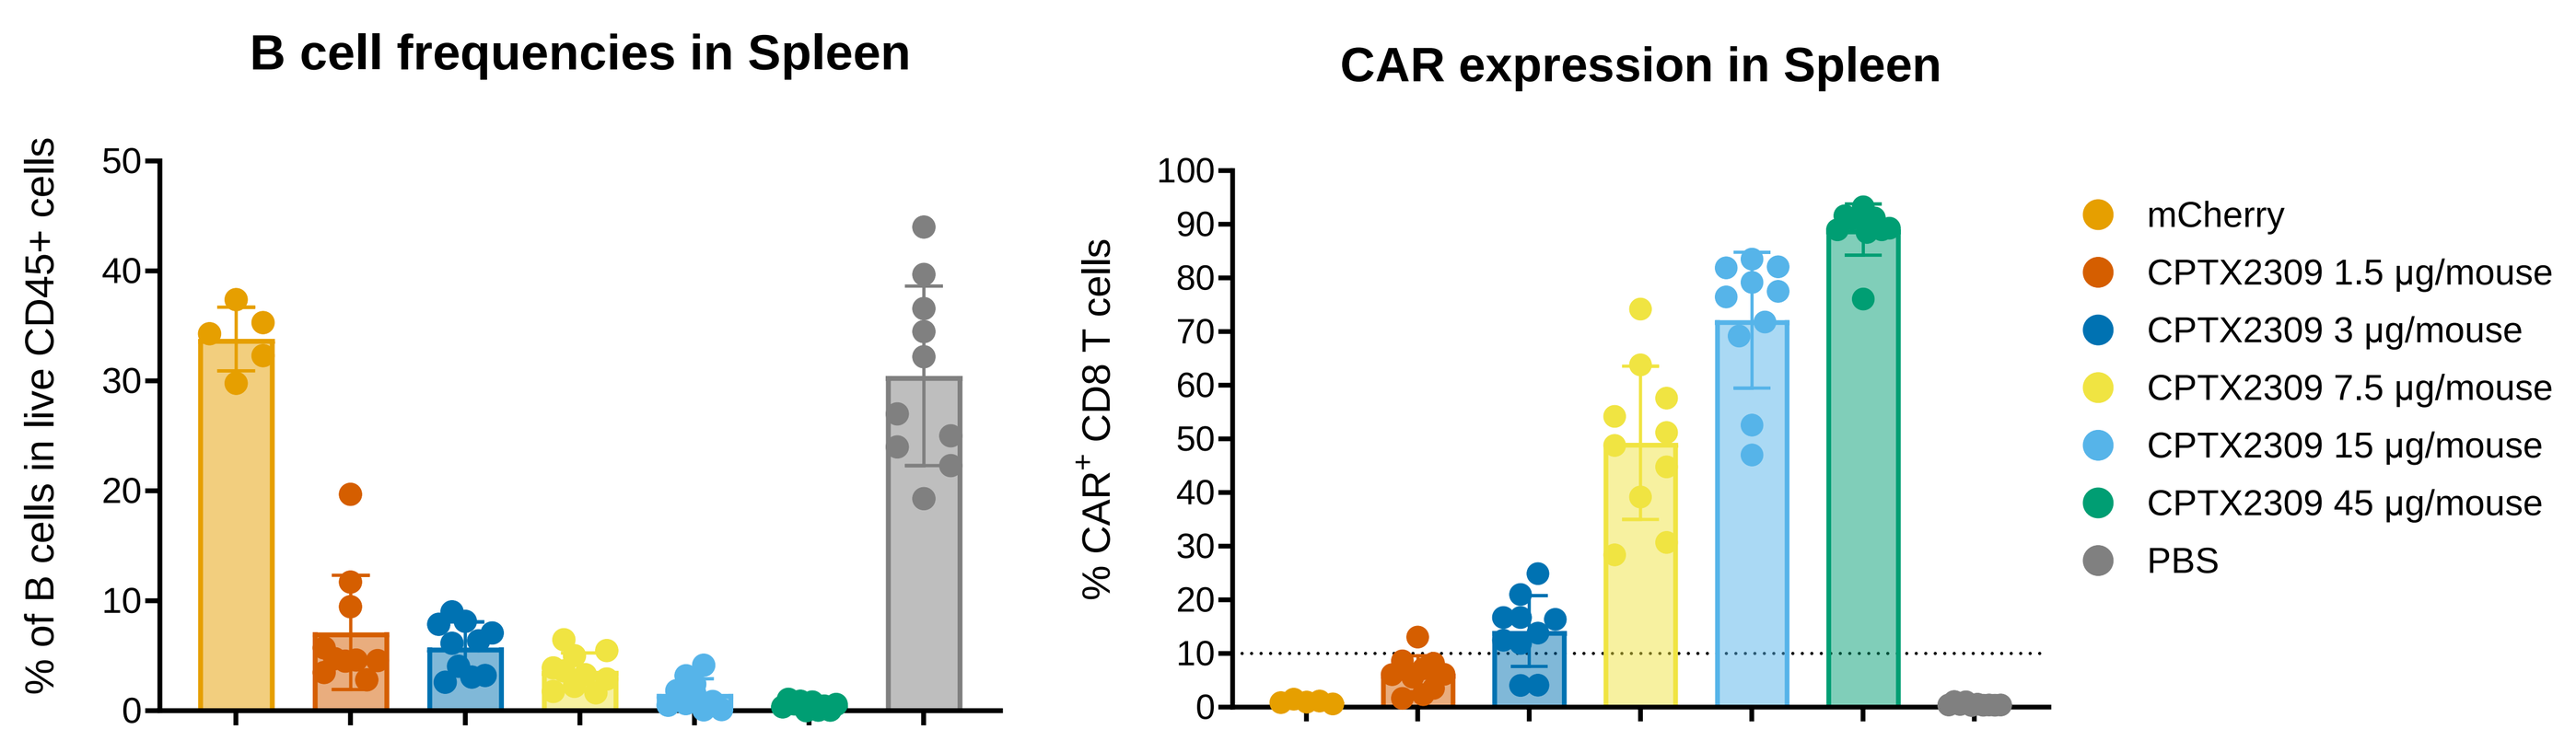
B cell frequencies in Spleen
% of B cells in live CD45+ cells
CAR expression in Spleen
% CAR+ CD8 T cells
mCherry
CPTX2309 1.5 µg/mouse
CPTX2309 3 µg/mouse
CPTX2309 7.5 µg/mouse
CPTX2309 15 µg/mouse
CPTX2309 45 µg/mouse
PBS

B cell frequencies (left) and CAR expression percentage in CD8 T cells (right) in NCG mice engrafted with human PBMCs 24 hours after dosing with indicated tLNP. Dose of mCherry control was 7.5 µg.

FIGURE 7 | CPTX2309 IS EFFECTIVE IN BOTH CD34 HUMANIZED MOUSE MODEL AND NALM6 TUMOR MODEL

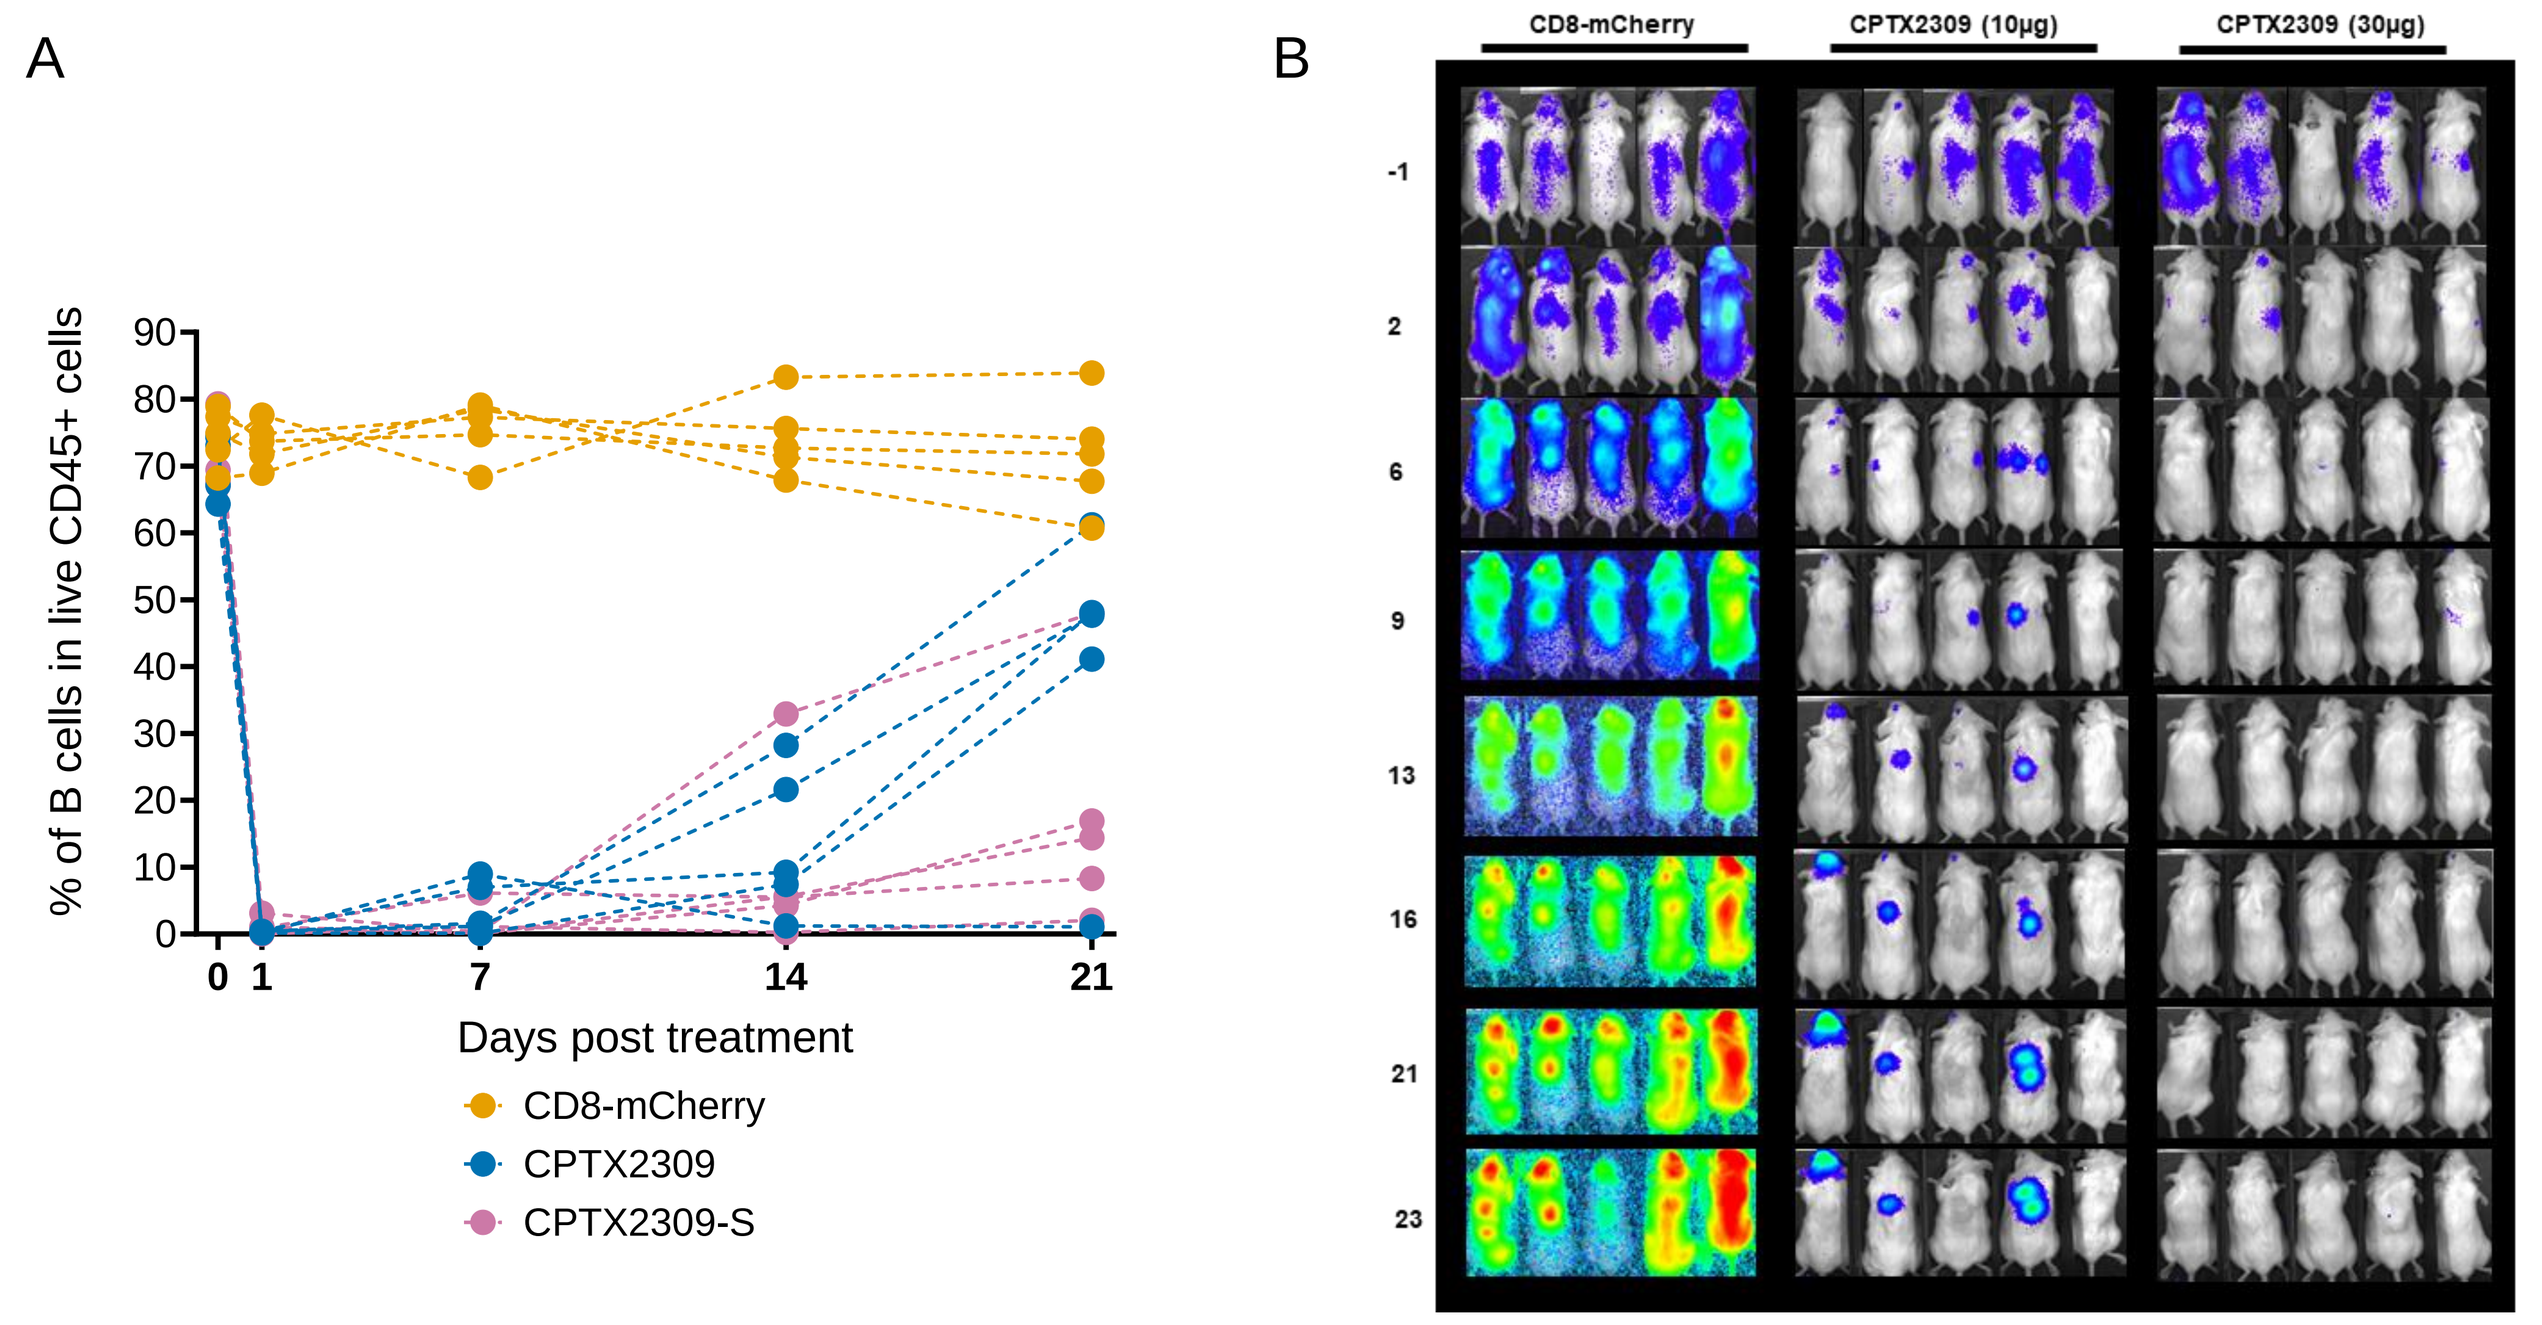
A
% of B cells in live CD45+ cells
Days post treatment
CD8-mCherry
CPTX2309
CPTX2309-S
B
CD8-mCherry
CPTX2309 (10µg)
CPTX2309 (30µg)

A) Time course of primary B cell percentage in NSG-CD34 mice dosed with 30 µg mCherry control tLNP, CPTX2309, or the same tLNP formulation containing an anti-CD20 mRNA CAR payload (CPTX2309-S). Mice were dosed every 3 days for 3 doses. B) NCG mice were engrafted with 1 x 10^6 human PBMCs on day -18 and inoculated with 5 x 10^5 Nalm6-luciferase tumor cells on day -8. Mice were bled and staged on day -1 based on tumor burden and human CD45+ cell engraftment. Mice were then dosed with indicated tLNP beginning on day 0, twice weekly for a total of 5 doses. Bioluminescence images at the indicated timepoint are shown.

CONCLUSION

CPTX2309 is a novel *in vivo* CAR therapy designed to bring together the unprecedented potency of CARs with the scalability and feasibility of the CellSeeker™ tLNP-mRNA platform. CPTX2309 rapidly, efficiently, and preferentially engineers anti-CD19 CAR CD8 T cells resulting in profound pharmacological activity both *in vitro* and *in vivo*, against primary or malignant human B cells. These data support the development of CPTX2309 for a broad range of B cell disorders, including autoimmune diseases, and point to the broad potential of this platform technology.

Reprinted from the ACR Convergence held November 2024. The American College of Rheumatology does not guarantee, warrant, or endorse any commercial products or services. Reprinted by Capstan Therapeutics, Inc.